Different cornea profile?

KEEP CALM AND FIT SPECIALTY SOFT LENS

Muzzi Maria Giulia, technical support manager Occhio x Occhio; Giulianelli Riccardo, optometrist - Italy

Introduction

Keratoconus is the classic case in which a RGP contact lens provides a new refracting surface for an irregular cornea and gives improved visual acuity, whereas spectacles frequently offer no signifcant benefts. Also in PRK, a laser technique used to sculpt the central corneal surface producing a reduction in myopia, RGP lenses are used after the operation(1). RGP lenses are considered the primary visual correction tool for irregular cornea, like keratoconus or post-surgery corneas. Even with the variety geometry, designs and options available, RGP lenses are often difficult to fit for some irregular corneas. Patients sometimes experiencing fluctuating vision, discomfort or RGP lens intolerance(2). To increase comfort, lens centration and visual acuity hybrid lenses and scleral lenses today present an effective solution for irregular cornea correction, but main problems are settling on the cornea and handling. In order to help fitters soft contact lenses for irregular cornea are available. Soft contact lenses can sometimes be used for irregular cornea, depending upon the degree of corneal distortion.The main purpose to use these type of lenses is to improve comfort and the easy handling that we can offer to the patients, though the visual acuity is not the same like RGP lenses or hybrid lenses. It is also important to know that these lenses cannot be used in every case of irregular cornea. Menicon offers Rose K2 Soft with an improved design available also in Silicon Hydrogel material, better if compared to the previous soft contact lens design for irregular cornea. This lens, with a standard diameter of 14.80 mm, is fitted considering the base curve which yields the best visual acuity, fitting as flat as possible. It is possible to perfect the fitting by choosing the peripheral fit to optimize lens fit, location and movement. It is important to note that the periphery of the lens can be adjusted independently of the base curve(3). The purpose of this poster is to show how a soft lens design for irregular cornea can help fitters to offer good solution to the patients, preserving ocular health, visual acuity and comfort.

Case #1: Bilateral Keratoconus

Patient: 25 year old, male

Diagnosis: Bilateral keratoconus, cross-linking 2 years ago

Occupation: Frequently outdoors, work 8-10 hours a day in office

CURRENT STATUS

Spectacles: RE: -1.00 -4.00 60° 20/40, LE: -0.75 -2.00 120° 20/40

Contact lens: Never used. The ophthalmologist discovers bilateral keratoconus and advises the use of contact lenses for irregular cornea.

Fitting: At the begin fitted with RGP Rose K2. RE Rose K2 Nipple Cone and LE Rose K2, due to the different location and stadium of the keratoconus. Fit well, good visual acuity (RE: 20/22, LE 20/22; Binocular vision 20/20)(4) and good tolerance. Symptoms of scratchiness and pain after 3/4 hours of wear. At the control with the slit lamp, we noticed corneal staining and beginning of corneal erosion. Topography shows changement in keratoconus form. Immediately stopped the use of contact lenses and after 1 month and a half cornea was perfectly recovered and the ophthalmologist gave consent to use contact lenses.

FITTING SEQUENCE ROSE K2 SOFT (RE)

Trials Lens	Fitting Characteristics
7.60 : 14.80 : -05.00 : LIFT 00.00 : EP 0.35	NO MOVEMENT, LASER MARK 6H STABLE
7.80 : 14.80 : -04.00 : LIFT 00.00 : EP 0.35	MOVEMENT 1MM, LASER MARK 6H STABLE
8.00 : 14.80 : -03.00 : LIFT 00.00 : EP 0.35	TOO MUCH MOVEMENT, LASER MARK NOT AT 6H

FITTING SEQUENCE ROSE K2 SOFT (LE)

Trials Lens	Fitting Characteristics
8.40 : 14.80 : -01.00 : LIFT 00.00 : EP 0.35	LENS BLOCKED, UNCOMFORTABLE, SCLERA INDENTATION
8.60 : 14.80 : +00.00 : LIFT 00.00 : EP 0.35	NO MOVEMENT, LASER MARK 6H STABLE
8.80 : 14.80 : +01.00 : LIFT 00.00 : EP 0.35	MOVEMENT 1.5/2.00MM, LASER MARK 6H NOT TO MUCH STABLE BUT GOOD

Comments: The patient was so impressed in comfort and easy handling. Reduce Edge Lift in the left lens to stabilize the mark laser at 6h. Increase central thickness from 0.35 to 0.40 to improve vision.

Over-refraction: RE +0.50 -4.50 30° (20/25)(4), LE -2.75 -1.25 130° (20/22)(4), Binocular vision 20/22(4)

Final RE Lens: Rose K2 Soft Toric 7.80 14.80 -03.50 -4.50 30° EL 00.00 EP 0.40

Final LE Lens: Rose K2 Soft Toric 8.80 14.80 -01.75 -1.25 130° EL -1.00 EP 0.40

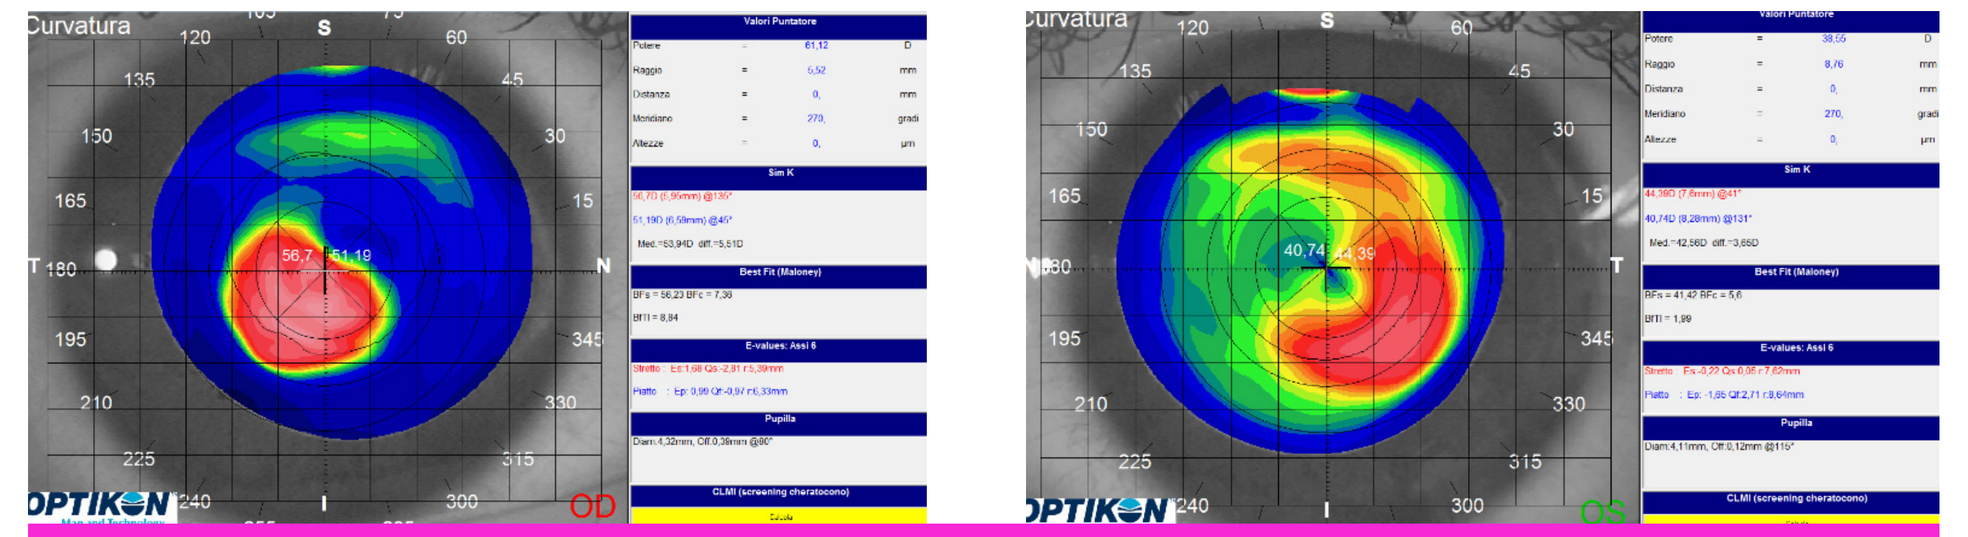
FIG. 1 KERATRON NOVA, CURVATURE TOPOGRAPHY TO BEGIN THE FITTING

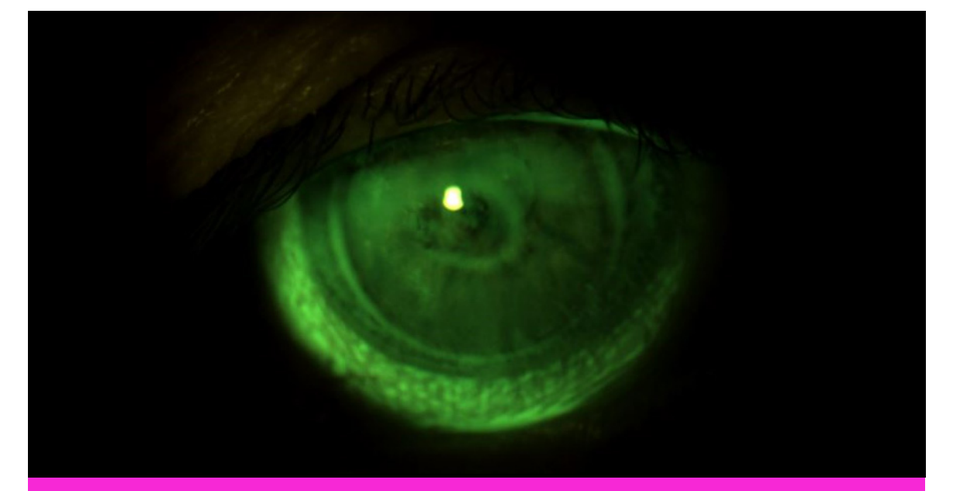
FIG. 2 RE. STAINING

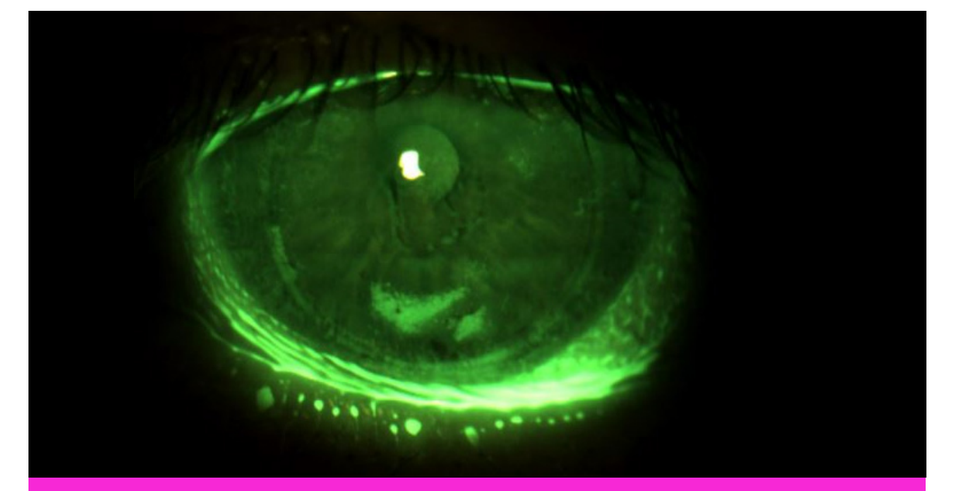
FIG.3 LE STAINING

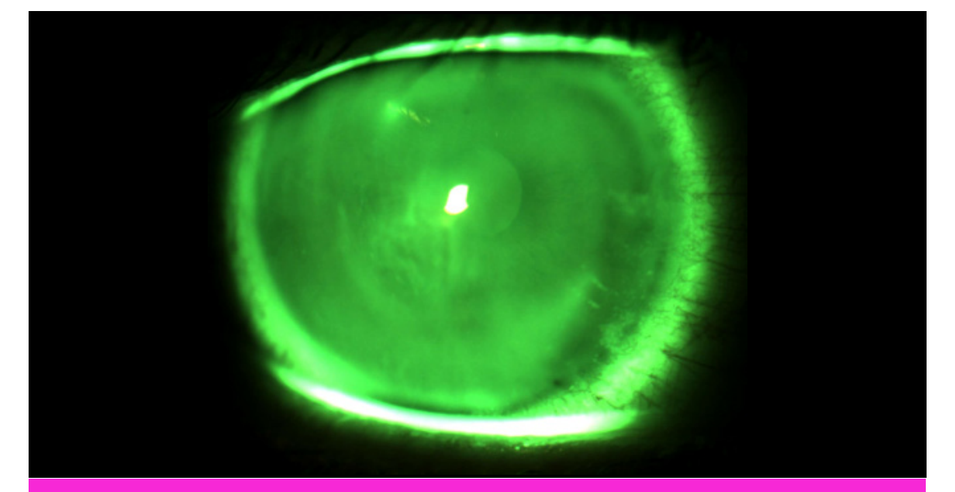
FIG. 4 RE CORNEA AFTER STOP USING RGP

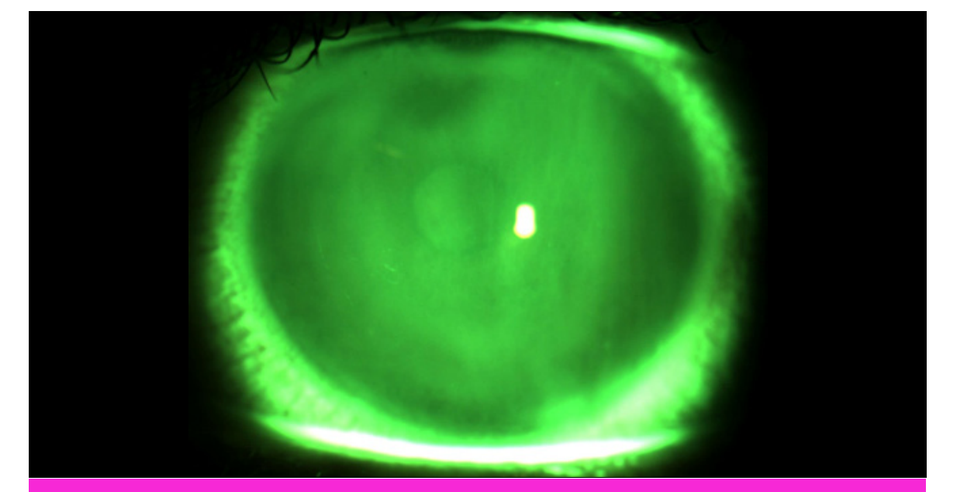
FIG. 5 LE CORNEA AFTER STOP USING RGP

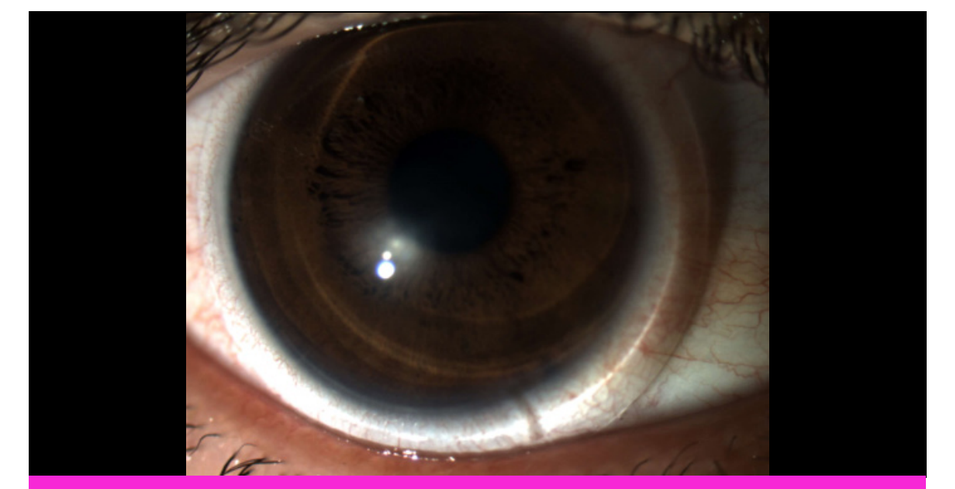
FIG. 6 RE ROSE K2-SOFT TORIC

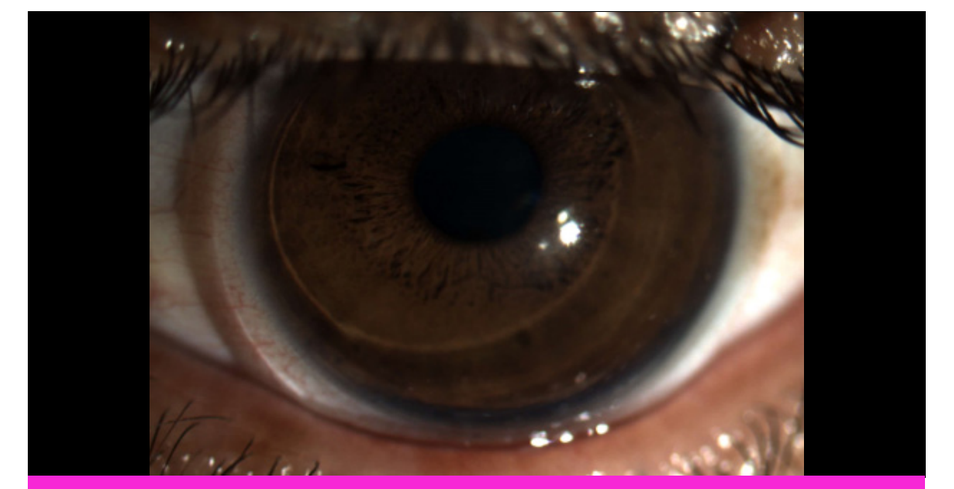
FIG. 7 LE ROSE K2-SOFT TORIC

Case #2: Post surgery cornea

Patient: 45 year old male

Diagnosis: Post Surgery cornea only on left eye. Operated three times, at the age of 20 and 25 years old for PRK and at the age of 43 for cataract and reducing double vision.

Occupation: Frequently outdoors, metalworker

CURRENT STATUS

Spectacles: +1.75 -0.75 144° (20/32)(4) He plains about double vision with his glasses.

Contact lens: Never used.

Other: Increase frontal and left side headaches over the last 20 month.

Fitting: At the begin fitted with RGP contact lens for post lasik cornea but the patient was not in comfort due to the fact that he wears the lens only in one eye. We try semi-scleral an hybrid contact lens but the patient refused this type of solution due to handling problems.

FITTING SEQUENCE ROSE K2 SOFT (LE)

Trials Lens	Fitting Characteristics
8.60 : 14.80 : +00.00 : LIFT 00.00 : EP 0.35	MOVEMENT 1MM, COMFORTABLE, GOOD VISION IMMEDIATELY AFTER THE BLINK, DOWN DECENTRALIZATION
8.80 : 14.80 : +01.00 : LIFT 00.00 : EP 0.35	MOVEMENT 1MM, COMFORTABLE, PERFECT VISION, AFTER 2 MINUTES DOWN DECENTRALIZATION , LASER MARK AT 6H

Comments: The patient was so impressed in comfort and easy handling. Reduce Edge lift in order to center the lens and avoid down decentralization due to cornea's form. Increase central thickness from 0.35 to 0.45 to improve vision and try to reduce double vision.

Over-refraction: LE -02.50, no cylindrical power was required on over refraction (20/22)(4)

Final LE Lens: Rose K2 Soft 8.80 14.80 -01.50 EL -1.00 EP 0.45 (20/22)(4)

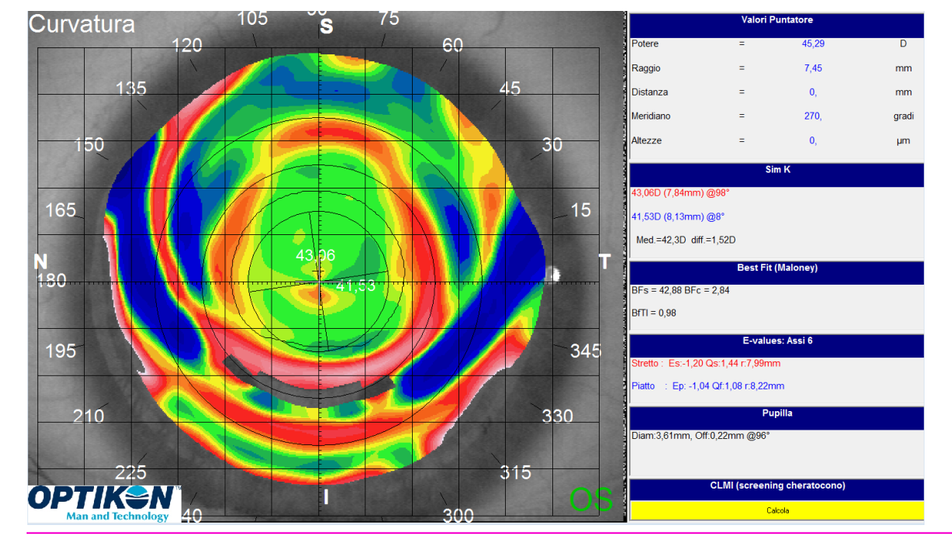
FIG.8 KERATRON NOVA, CURVATURE TOPOGRAPHY TO BEGIN THE FITTING

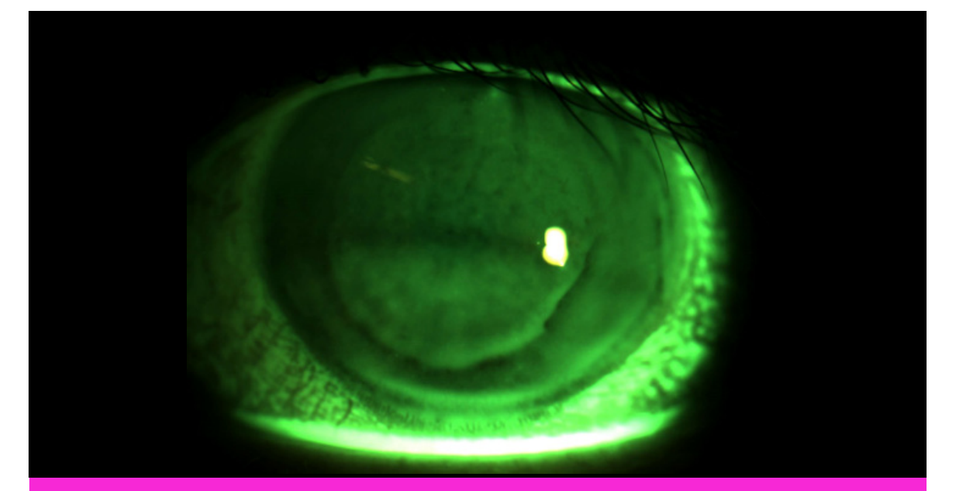
FIG. 9 LE CORNEA WITHOUT CONTACT LENS

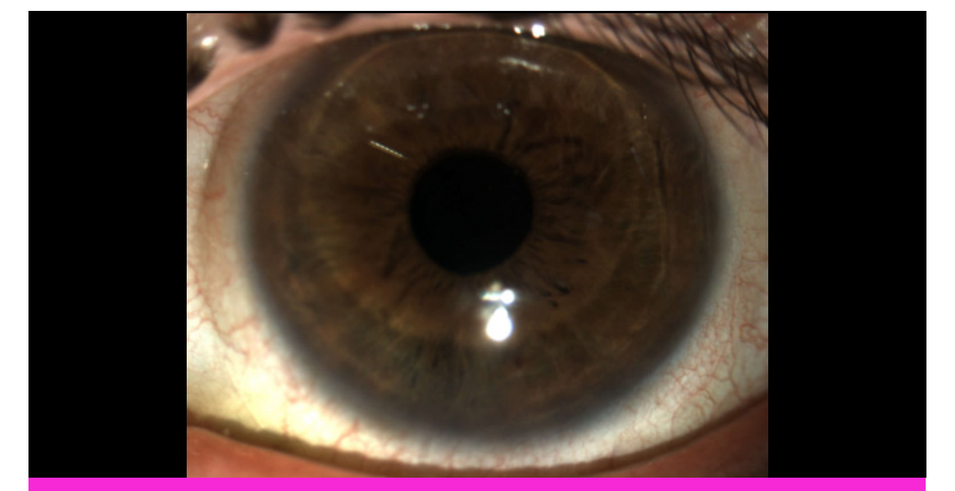
FIG.10 LE WITH ROSE K2-SOFT TORIC

Conclusion

Patient n°1 developed an intolerance in RGP contact lenses due to the sudden evolution of the keratoconus but he would not stop to use contact lens because his vision is better than spectacles. Soft contact lenses for irregular cornea were fitted successfully on both eyes and the patient uses his contact lenses for 10/ 12 hours. Patient n°2 presented double vision and complaining of visual difficulties especially at night. He noticed also an increase of headaches. In three weeks, there has been complete resolution of his headaches. Central thickness increased helps him to reduce his double vision. Booth patients are extremely in comfort and impressed in easy handling. Having motivation and good support can help patients with irregular cornea to achieve better vision than they could imagine. Patients could be skeptical at the beginning but practitioner have only to show them what they can obtain thanks to the contact lenses. After proving that their vision can improve with very comfortable lenses, these patients will continue to work closely with their optometrist following every advice and indications(5).

References

1. Andrew Gasson and Judith A. Morris, The Contact Lens Manual – A practical guide to fitting, Butterworth Heinemann, Elsevier
2. Downie, L.E. et al. Contact lens management ok keratoconus. Clin EXP Optom. 2015; 98: 299-311
3. Menicon Rose K2-Soft fitting guide. Available at: www.roseklens.com
4. Visual Acuity Snellen by feet
5. Manual of Contact Lens Prescribing and fitting with CD Rom, Milton M. Hom. (2000) Butterworth Heinemann